Abuse of XTAMPZA® ER Relative to Other Opioid Analgesics

Severtson, SG[1], Olsen, H[1], Iwanicki, JL[1], **Dart, RC**[1] [1]*Rocky Mountain Poison & Drug Safety - Denver Health, Denver, CO*

INTRODUCTION

- Extended-release (ER) oxycodone products are schedule II opioid analgesics designed to provide pain relief for a longer period of time than immediate-release (IR) oxycodone.
- Some individuals who abuse ER oxycodone tamper with the medication in order to bypass the controlled-release mechanism to release the entire amount of drug in a short period of time.
- XTAMPZA® ER is an abuse deterrent ER oxycodone product that uses DETERx® technology designed to discourage manipulation for the purposes of abuse. Studies suggest that XTAMPZA ER maintains ER properties after physical manipulation and administration via the oral or nasal routes.
- The FDA has granted XTAMPZA ER abuse-deterrent labeling with respect to oral, nasal, and intravenous routes of administration.
- We compare rates of abuse and misuse of XTAMPZA ER to other abuse deterrent formulation (ADF) ER opioids and IR oxycodone.

METHODS

- Data from 2018-2019 were analyzed from two Researched Abuse, Diversion and Addiction-Related Surveillance (RADARS®) System data sources:
- <u>Poison Center Program:</u> Rates of intentional abuse/misuse/unknown exposures were compared from 53 of 55 US poison centers
- <u>Treatment Center Programs Combined:</u> Odds of past month abuse prevalence adjusted for drug utilization were compared among respondents entering treatment for opioid use disorders
- XTAMPZA ER was compared to IR oxycodone and other ADF ER opioids.
- Drug utilization data (prescription and morphine equivalent grams dispensed) were from the IQVIA® (Danbury, CT) US-Based Longitudinal Patient Data.
- Missing values within opioid molecule were imputed across 50 data sets. Analyses were performed in each of the 50 data sets and estimates averaged.

Disclosure

The RADARS® System is supported by subscriptions from pharmaceutical manufacturers, government and non-government agencies for surveillance, research and reporting services. RADARS System is the property of Denver Health and Hospital Authority, a political subdivision of the State of Colorado. Denver Health retains exclusive ownership of all data, databases and systems. Subscribers do not participate in data collection nor do they have access to the raw data. This research was sponsored by Collegium Pharmaceutical. XTAMPZA® ER is a registered trademark of Collegium Pharmaceutical.

Table 1. Prescriptions Dispensed by Drug Group, 2018-2019

Product Name	Number of Prescriptions	Percentage of Group
XTAMPZA® ER	737,565	100%
Other ADF ER Opioid Products	4,813,789	100%
OxyContin®	4,108,456	85.3%
Hysingla® ER	359,014	7.5%
Embeda®	253,677	5.3%
Morphabond® ER	76,901	1.6%
Arymo® ER	15,741	0.3%
IR Oxycodone	79,644,518	100%
IR Single Ingredient Oxycodone	32,130,814	40%
IR Combination Ingredient Oxycodone	47,513,704	60%

Figure 1. Ratio of Rates Relative to XTAMPZA ER by Drug Group with 95% CI, RADARS® System Poison Center Program, 2018 – 2019

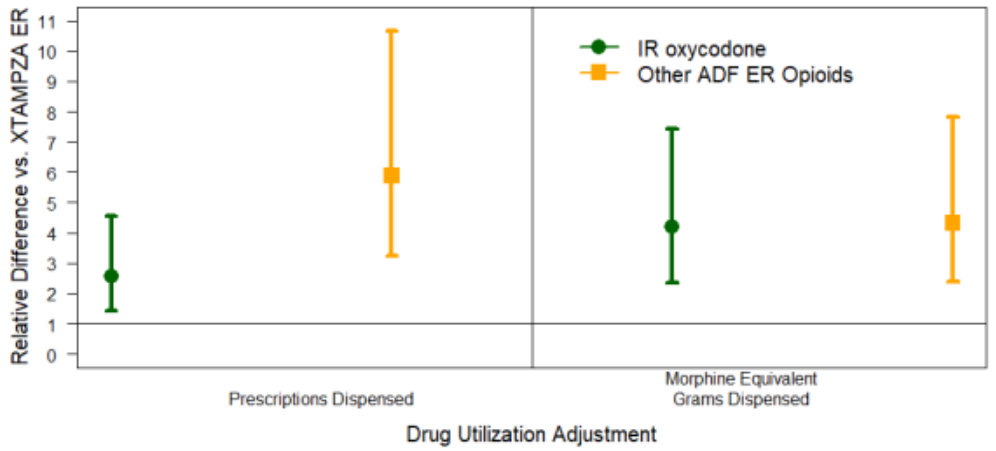
Relative Difference vs. XTAMPZA ER
IR oxycodone
Other ADF ER Opioids
Prescriptions Dispensed
Morphine Equivalent Grams Dispensed
Drug Utilization Adjustment

Figure 2. Ratio of Adjusted Prevalence Relative to XTAMPZA ER by Drug Group with 95% CI, , RADARS® System Treatment Center Programs Combined, 2018 – 2019

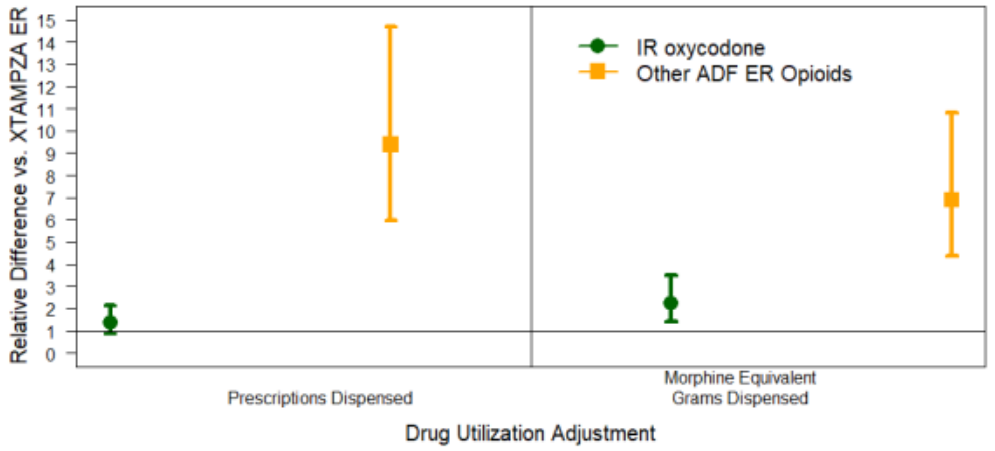
Relative Difference vs. XTAMPZA ER
IR oxycodone
Other ADF ER Opioids
Prescriptions Dispensed
Morphine Equivalent Grams Dispensed
Drug Utilization Adjustment

RESULTS

- There were 737,565 (increasing by quarter) prescriptions for XTAMPZA ER, 4.8 million (decreasing by quarter) for other ADF ER opioids, and 79 million (decreasing by quarter) for IR oxycodone (Table 1).
- Poison Center Program post-imputation rate ratios relative to XTAMPZA ER:
- Adjusting for prescriptions dispensed (Figure 1)
 - IR oxycodone rate ratio=2.55 (95% CI: 1.43 to 4.54, p=0.002)
 - Other ADF ER opioids rate ratio=5.89 (95% CI: 3.25-10.67, p<0.001)
- Adjusting for morphine equivalent grams dispensed (Figure 1)
 - IR oxycodone RR=4.19 (95% CI: 2.35-7.45, p<0.001)
 - Other ADF ER opioids RR= 4.33 (95% CI: 2.39-7.84, p<0.001)
- None of the 30 XTAMPZA ER cases involved injection or inhalation use
- Treatment Center Programs Combined post-imputation odds ratios (OR) relative to XTAMPZA ER:
- Adjusted for prescriptions dispensed (Figure 2):
 - IR oxycodone OR= 1.37 (95% CI: 0.88-2.14, p=0.162)
 - Other ADF ER opioids OR=9.38 (95% CI: 5.99-14.71, p<0.001)
- Adjusted for morphine equivalent grams dispensed (Figure 2)
 - IR oxycodone OR=2.22 (95% CI: 1.42-3.48, p<0.001)
 - Other ADF ER opioids OR=6.88 (95% CI: 4.39-10.79, p<0.001)
- Of the 21 XTAMPZA ER abuse cases, 2 endorsed injection use, 2 endorsed inhalation use.

CONCLUSIONS

- Abuse and misuse exposures involving XTAMPZA ER reported to poison centers and XTAMPZA ER abuse cases among individual entering treatment for opioid use disorders were infrequent relative to comparators.
- No abuse or misuse of XTAMPZA ER via unintended routes of administration was reported in the Poison Center Program.

LIMITATIONS

- Abuse of specific products is self-reported.
- In the Treatment Center Programs Combined, even if false positive endorsements on the survey are rare they are more likely to disproportionately inflate estimates of less common products such as XTAMPZA ER than more commonly prescribed comparators.
- This analysis is limited to a specific time periodic and could change over time.