The Diversion and Street Price of XTAMPZA® ER Relative to Other Prescription Opioids

Severtson, SG[1], Margolin, ZR[1], May, K[1], Iwanicki, JL[1], **Dart, RC**[1] [1]*Rocky Mountain Poison & Drug Safety - Denver Health, Denver, CO*

ROCKY MOUNTAIN POISON & DRUG SAFETY
Saving lives with answers.™
RADARS® SYSTEM

INTRODUCTION

- Many individuals who abuse prescription opioids obtain these medications after they are diverted from legal distribution channels.
- The frequency with which a particular product is diverted and the price paid in illegal markets may be indicators of the demand among individuals who abuse opioids.
- XTAMPZA® ER is an abuse deterrent extended-release (ER) oxycodone product that uses DETERx® technology designed to discourage product manipulation for misuse and abuse by oral and non-oral routes.
- This study assesses the extent of diversion and average street price paid for XTAMPZA ER relative to other prescription opioid analgesics from 2018-2019.

METHODS

- Data collected from 2018 through 2019 were analyzed from two Researched Abuse, Diversion and Addiction-Related Surveillance (RADARS®) System data sources:
- **Drug Diversion Program-** Information on new cases of diversion reported by law enforcement and regulatory agencies in 49 states
- **StreetRx Program-** black market drug price information via anonymous submissions to the StreetRx.com website
- XTAMPZA ER was compared to immediate-release (IR) oxycodone and other abuse deterrent formulation (ADF) ER opioids.
- With Drug Diversion Program data, rates of diversion cases were calculated per prescriptions dispensed and morphine milligram equivalent (MME) grams dispensed and compared. Both estimates were obtained from the IQVIA® (Danbury, CT) US-Based Longitudinal Patient Data.
- In the StreetRx Program, the geometric mean price per milligram strength was compared between drug groups. Unadjusted geometric mean street price and geometric mean street price adjusted for tablet strength and active pharmaceutical ingredient were assessed.

DISCLOSURE

The RADARS® System is supported by subscriptions from pharmaceutical manufacturers, government and non-government agencies for surveillance, research and reporting services. RADARS System is the property of Denver Health and Hospital Authority, a political subdivision of the State of Colorado. Denver Health retains exclusive ownership of all data, databases and systems. Subscribers do not participate in data collection nor do they have access to the raw data. This research was sponsored by Collegium Pharmaceutical. XTAMPZA® ER is a registered trademark of Collegium Pharmaceutical.

Table 1. Prescriptions Dispensed by Drug Group, 2018-2019

Product Name	Number of Prescriptions	Percentage of Group
XTAMPZA® ER	737,565	100%
Other ADF ER Opioid Products	4,813,789	100%
OxyContin®	4,108,456	85.3%
Hysingla® ER	359,014	7.5%
Embeda®	253,677	5.3%
Morphabond® ER	76,901	1.6%
Arymo® ER	15,741	0.3%
IR Oxycodone	79,644,518	100%
IR Single Ingredient Oxycodone	32,130,814	40%
IR Combination Ingredient Oxycodone	47,513,704	60%

Figure 1. Ratio of Rates Relative to XTAMPZA ER by Drug Group with 95% CI, RADARS® System Drug Diversion Program, 2018 – 2019

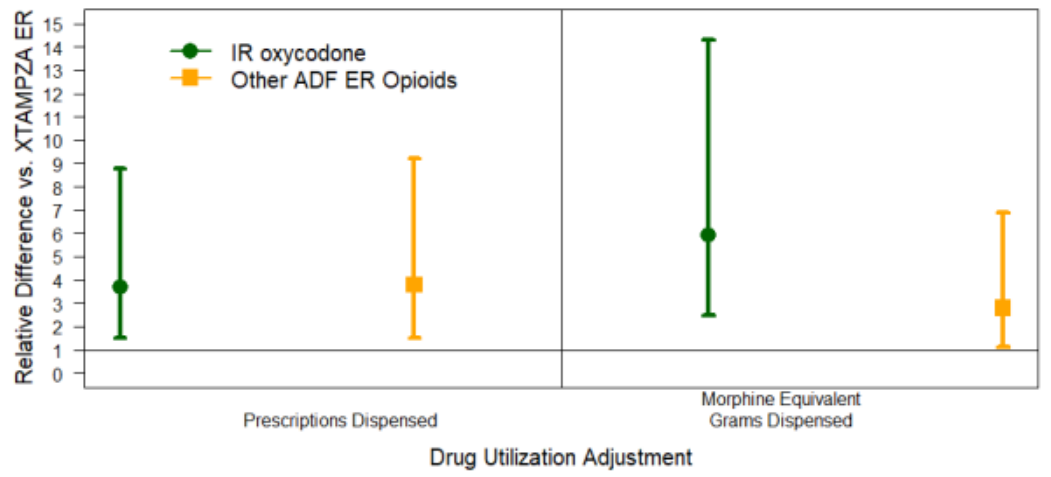
Relative Difference vs. XTAMPZA ER
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15
IR oxycodone
Other ADF ER Opioids
Prescriptions Dispensed
Morphine Equivalent Grams Dispensed
Drug Utilization Adjustment

Table 2. Unadjusted Geometric Mean Price Per mg by Drug Group, RADARS® System StreetRx Program, 2018 – 2019

Drug Group	Unadjusted Geometric Mean Price per mg (95% CI)
XTAMPZA® ER	$0.59 ($0.50 - $0.69)
Other ADF ER Opioid Products	$0.50 ($0.46 - $0.53)
IR Oxycodone	$1.04 ($1.00 - $2.07)

RESULTS

- There were 737,565 (increasing by quarter) prescriptions for XTAMPZA ER, 4.8 million (decreasing by quarter) for other ADF ER opioids, and 79 million (decreasing by quarter) for IR oxycodone (Table 1).
- There were 2,416 cases of diversion involving drug groups of interest; 5 involved XTAMPZA ER, 128 involved other ADF ER opioids, and 2,083 involved IR oxycodone. Rate ratios relative to XTAMPZA ER (Figure 1) were:
 - Adjusted prescriptions dispensed, the IR oxycodone diversion rate was 3.7 (95% CI: 1.5-8.8, p=0.004) times greater and the other ADF ER opioids diversion rate was 3.8 (95% CI: 1.5-9.2, p=0.004) times greater
 - Adjusted for the MME grams dispensed, the IR oxycodone diversion was 5.9 (95% CI: 2.5-14.3, p<0.001) times greater and the other ADF opioids diversion rate 2.8 (95% CI:1.1-6.9, p=0.024) times greater.
- Unadjusted geometric mean price of IR oxycodone was highest ($1.04 per mg, 95% CI: $1.00-$1.07), followed by XTAMPZA ER ($0.59, 95% CI: $0.50-$0.69), and other ADF ER Opioids ($0.50, 95% CI: $0.46-$0.53) (Table 2).
- After adjusting for tablet strength and active pharmaceutical ingredients, XTAMPZA ER had the lowest estimates price per milligram but differences with comparator drug groups were not statistically significant.

CONCLUSIONS

- Diversion of XTAMPZA ER was observed though it comprised a smaller number of cases than would be expected based on prescription volume.
- The appropriate comparator based on FDA guidelines is IR oxycodone. IR oxycodone had the highest number of cases and highest street price.
- Using two different drug availability measures, diversion of IR oxycodone and other ADF ER opioids was significantly higher than XTAMPZA ER.
- The street price of XTAMPZA ER was lower than IR oxycodone products. No differences in price per milligram were observed between XTAMPZA ER and comparator drug groups after adjusting for differences in tablet strength and active pharmaceutical ingredient.
- Though diversion of XTAMPZA ER is rare relative to other opioids, continued monitoring is warranted to identify sources and reasons for illegal acquisition.

LIMITATIONS

- The Drug Diversion Program is not a representative sample and may be affected by regional differences in law enforcement efforts.
- StreetRx Program data are self-report and subject to recall/recognition bias.
- These results look at a specific period of time and may change over time when more data is available.